SIRPα-Deficient CAR-Macrophages Exhibit Enhanced Anti-Tumor Function and Bypass the CD47 Immune Checkpoint

Chris Sloas[1], Rashid Gabbasov[1], Nicholas R. Anderson[1], Sascha Abramson[1], Michael Klichinsky[1], Yumi Ohtani[1]
[1]Carisma Therapeutics, Philadelphia, PA

INTRODUCTION

Background: CAR-Macrophages for targeting solid tumors

- CAR-Macrophage (CAR-M) technologies harness macrophage effector functions to attack solid tumors [1-3]

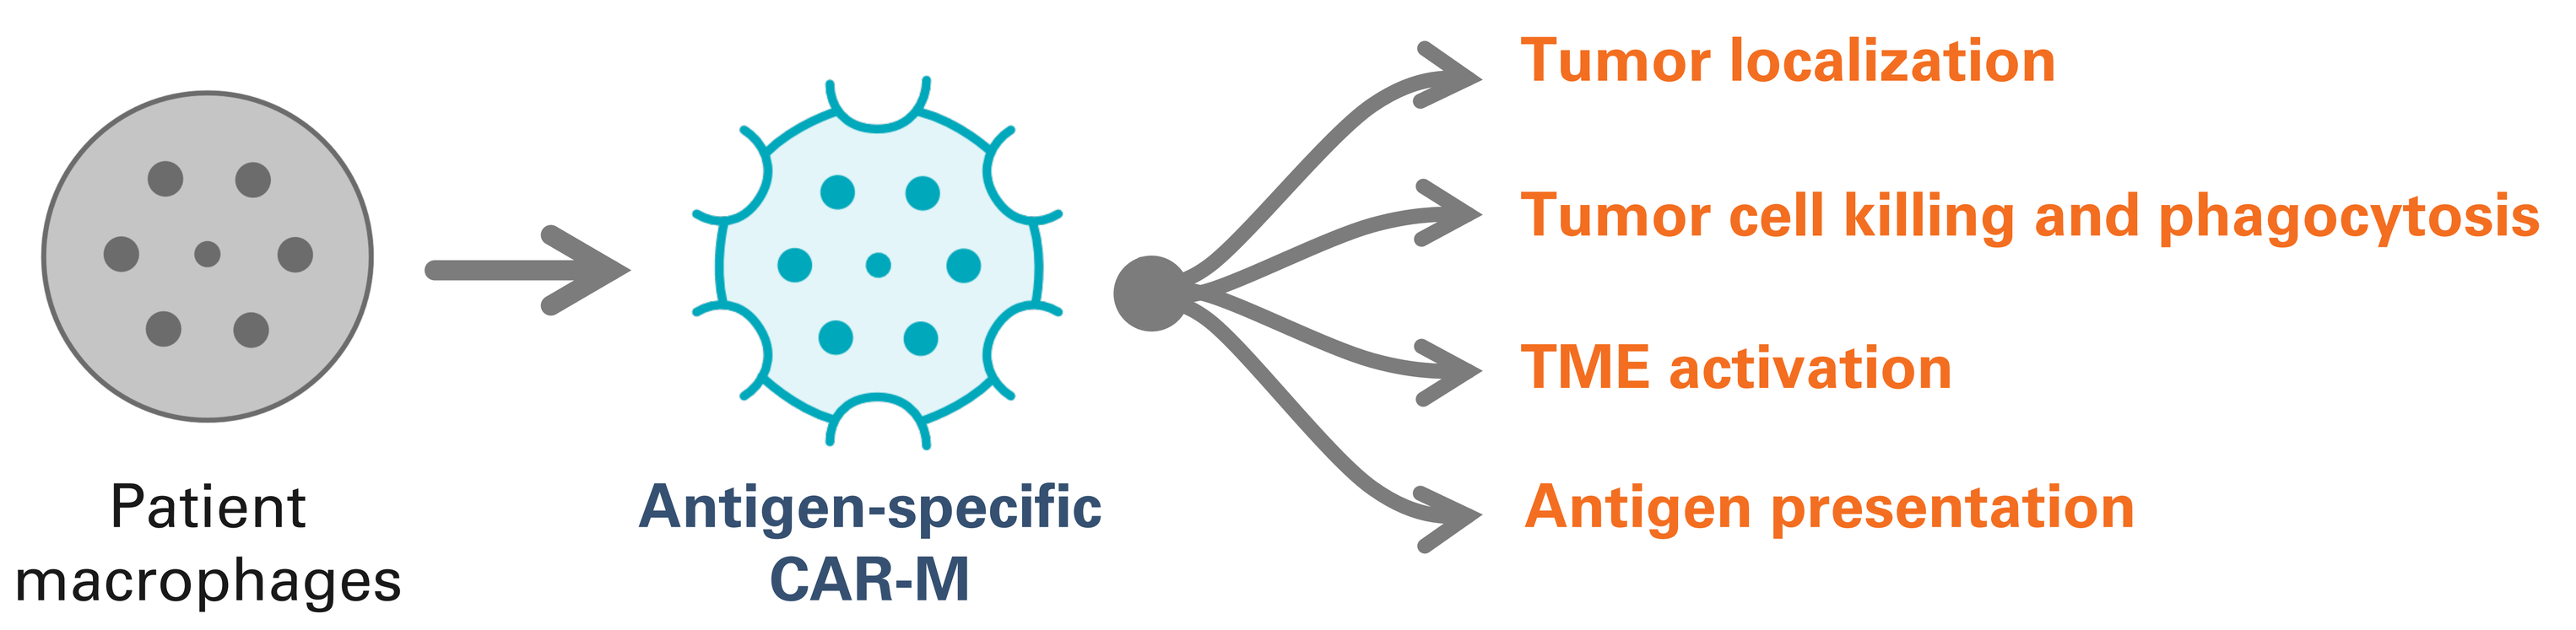
Patient macrophages
Antigen-specific CAR-M
Tumor localization
Tumor cell killing and phagocytosis
TME activation
Antigen presentation

CD47 reduces CAR-M phagocytosis, cytokine release, and can be reversed by CD47 blockade

- Tumors overexpress CD47 to evade phagocytosis by sending a "don't eat me" signal to macrophage-expressed SIRPα [4]

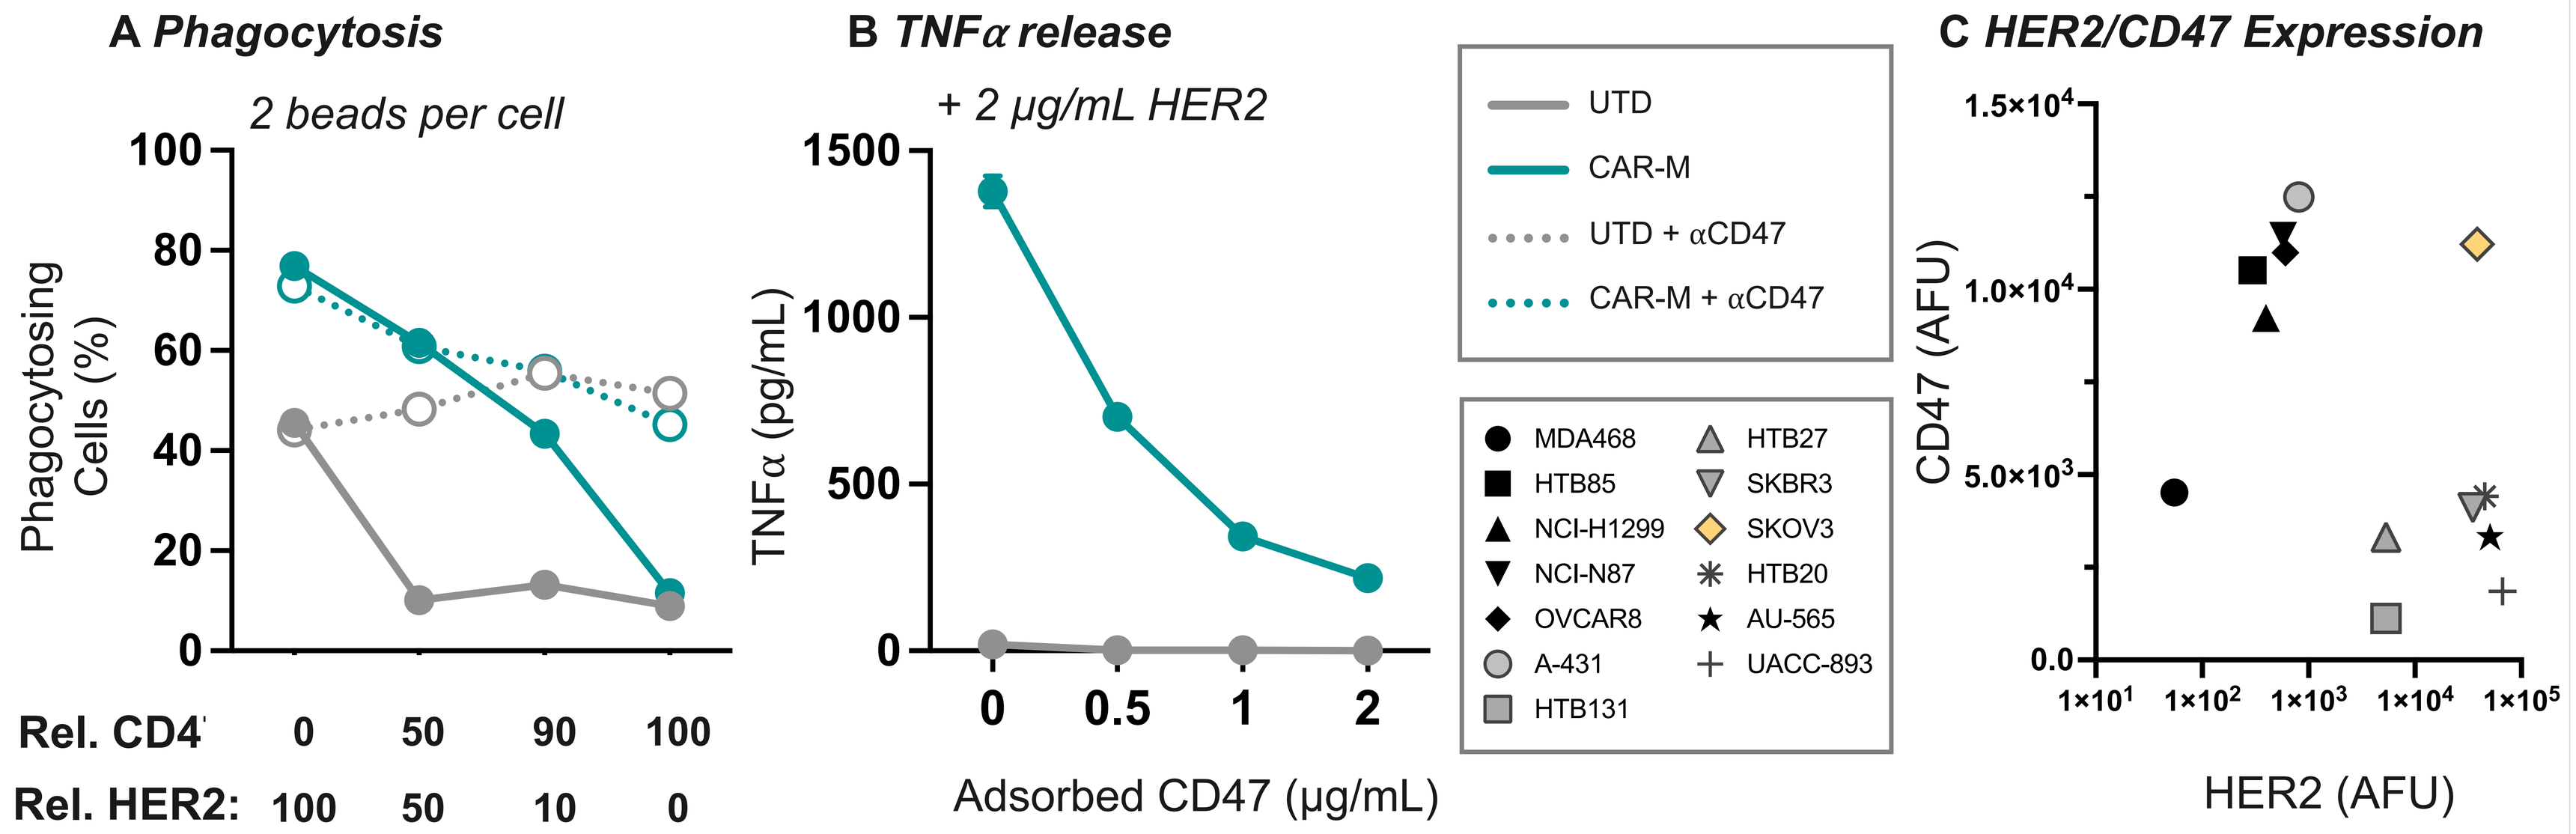
A Phagocytosis
2 beads per cell
Phagocytosing Cells (%)
Rel. CD4 0 50 90 100
Rel. HER2: 100 50 10 0
B TNFα release
+ 2 µg/mL HER2
TNFα (pg/mL)
Adsorbed CD47 (µg/mL)
UTD
CAR-M
UTD + αCD47
CAR-M + αCD47
C HER2/CD47 Expression
CD47 (AFU)
HER2 (AFU)
MDA468
HTB85
NCI-H1299
NCI-N87
OVCAR8
A-431
HTB131
HTB27
SKBR3
SKOV3
HTB20
AU-565
UACC-893

Figure 1. CD47 can inhibit antigen-induced CAR-M activity. (**A**) Phagocytosis of beads coated with varied ratios of HER2:CD47, measured by flow cytometry. (**B**) TNFα released from macrophages grown on surfaces coated with HER2 and CD47. CD47 inhibits CAR-M phagocytosis and TNFα release. CD47 blockade can rescue CAR-M phagocytosis. (**C**) SKOV-3 highly express HER2, CD47.

Goal: Generate SIRPα knockout primary human CAR-M that resist CD47-inhibition

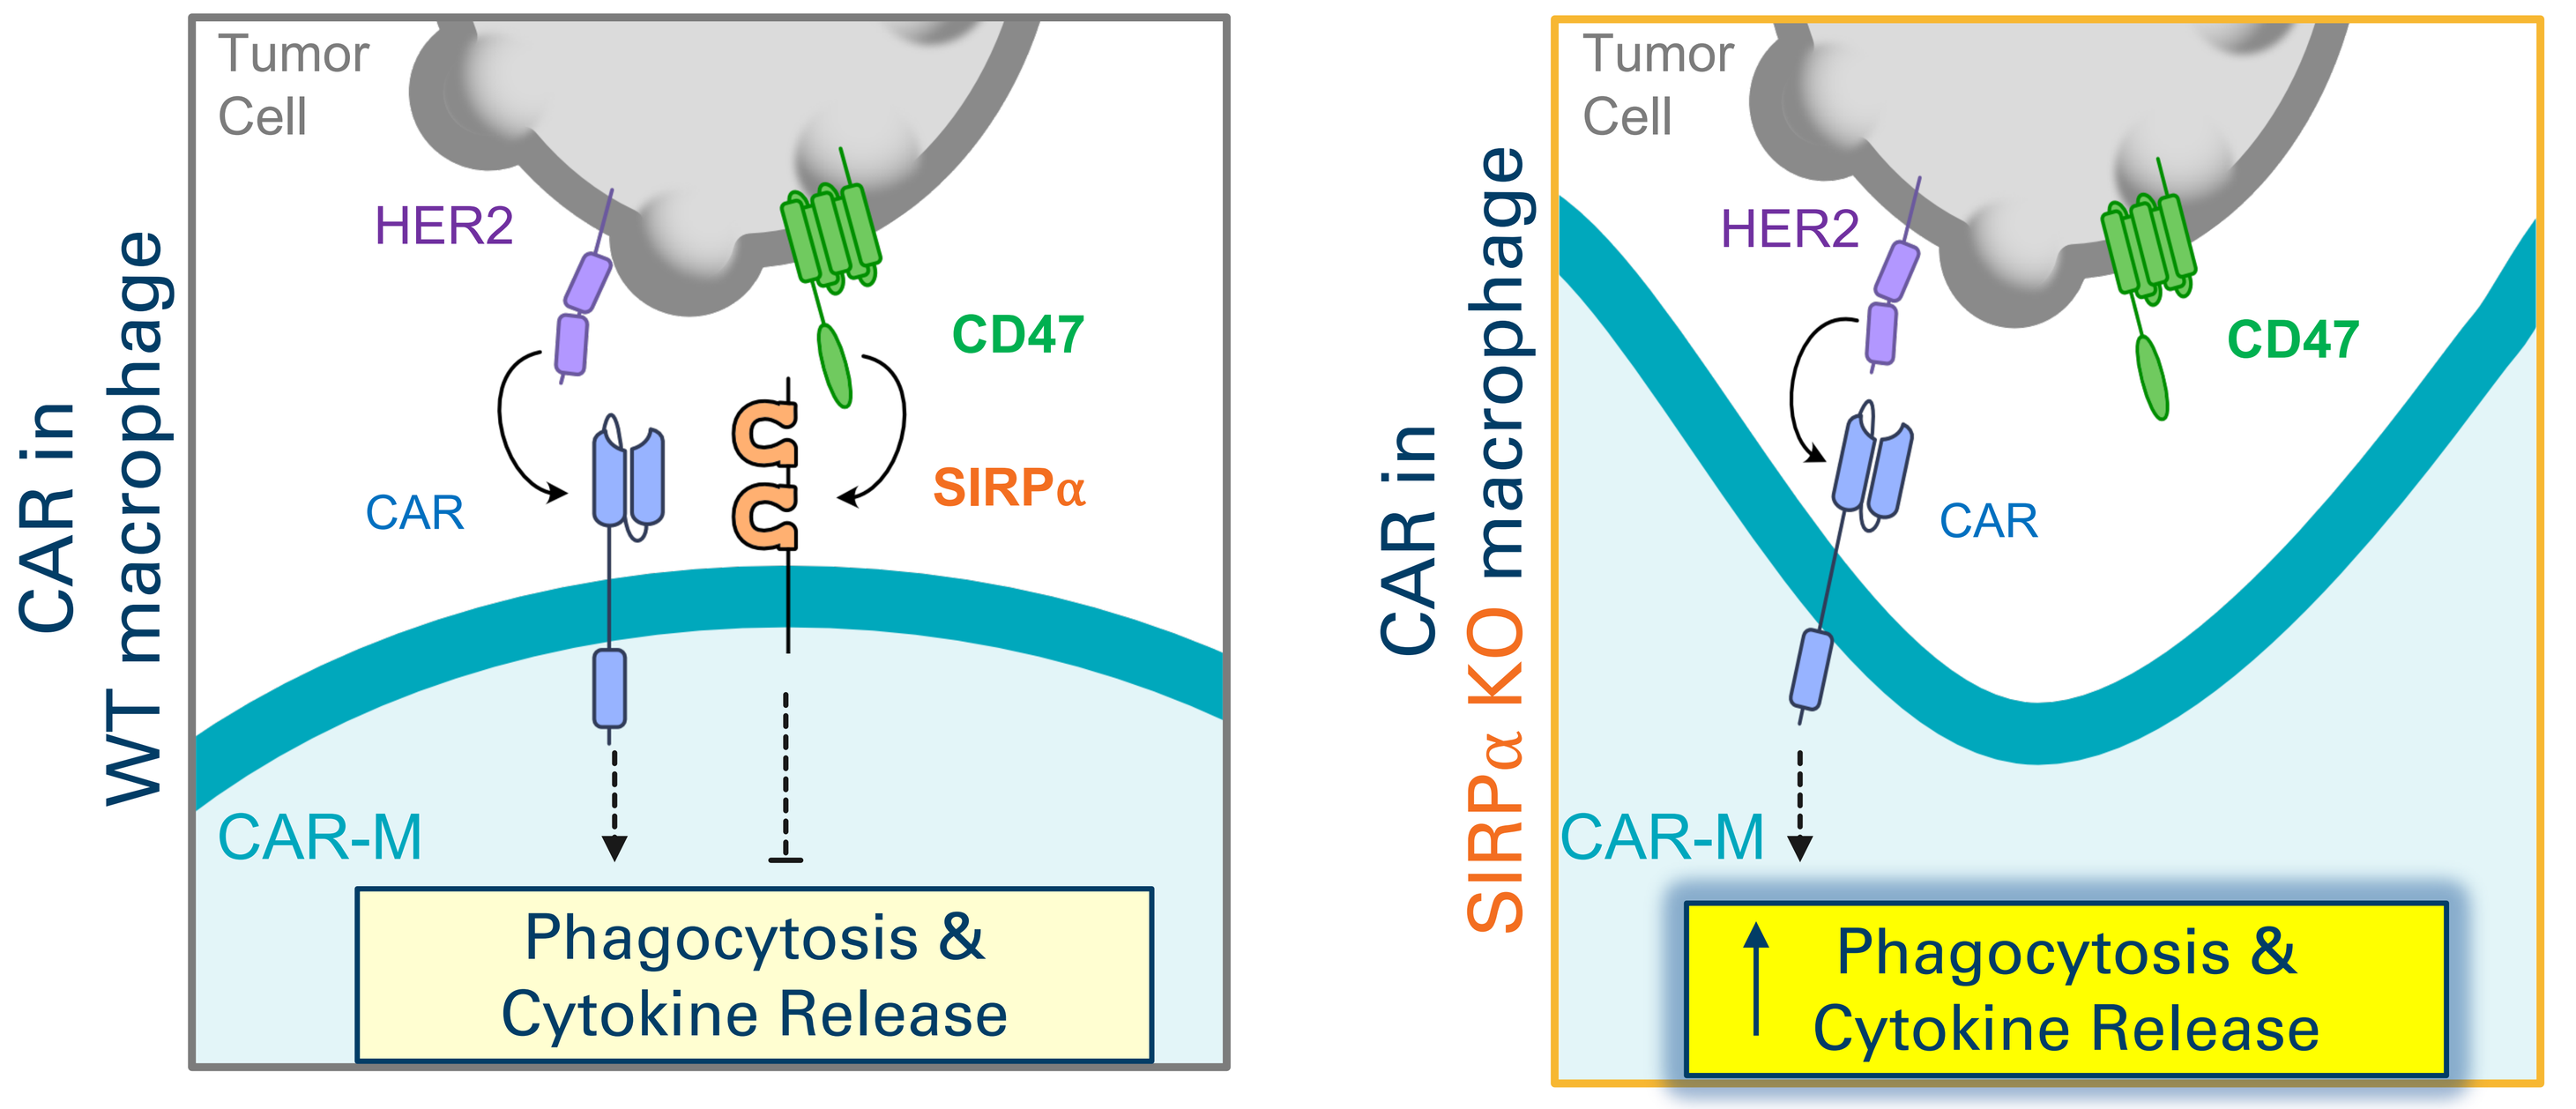
CAR in WT macrophage
Tumor Cell
HER2
CD47
CAR
SIRPα
CAR-M
Phagocytosis & Cytokine Release
CAR in SIRPα KO macrophage
Tumor Cell
HER2
CD47
CAR
CAR-M
↑ Phagocytosis & Cytokine Release

CRISPR/Cas9 was utilized to generate SIRPα knockout primary human control or anti-HER2 CAR-M

ANTI-TUMOR FUNCTION OF SIRPα-KO CAR-M

SIRPα is efficiently knocked out in primary human macrophages

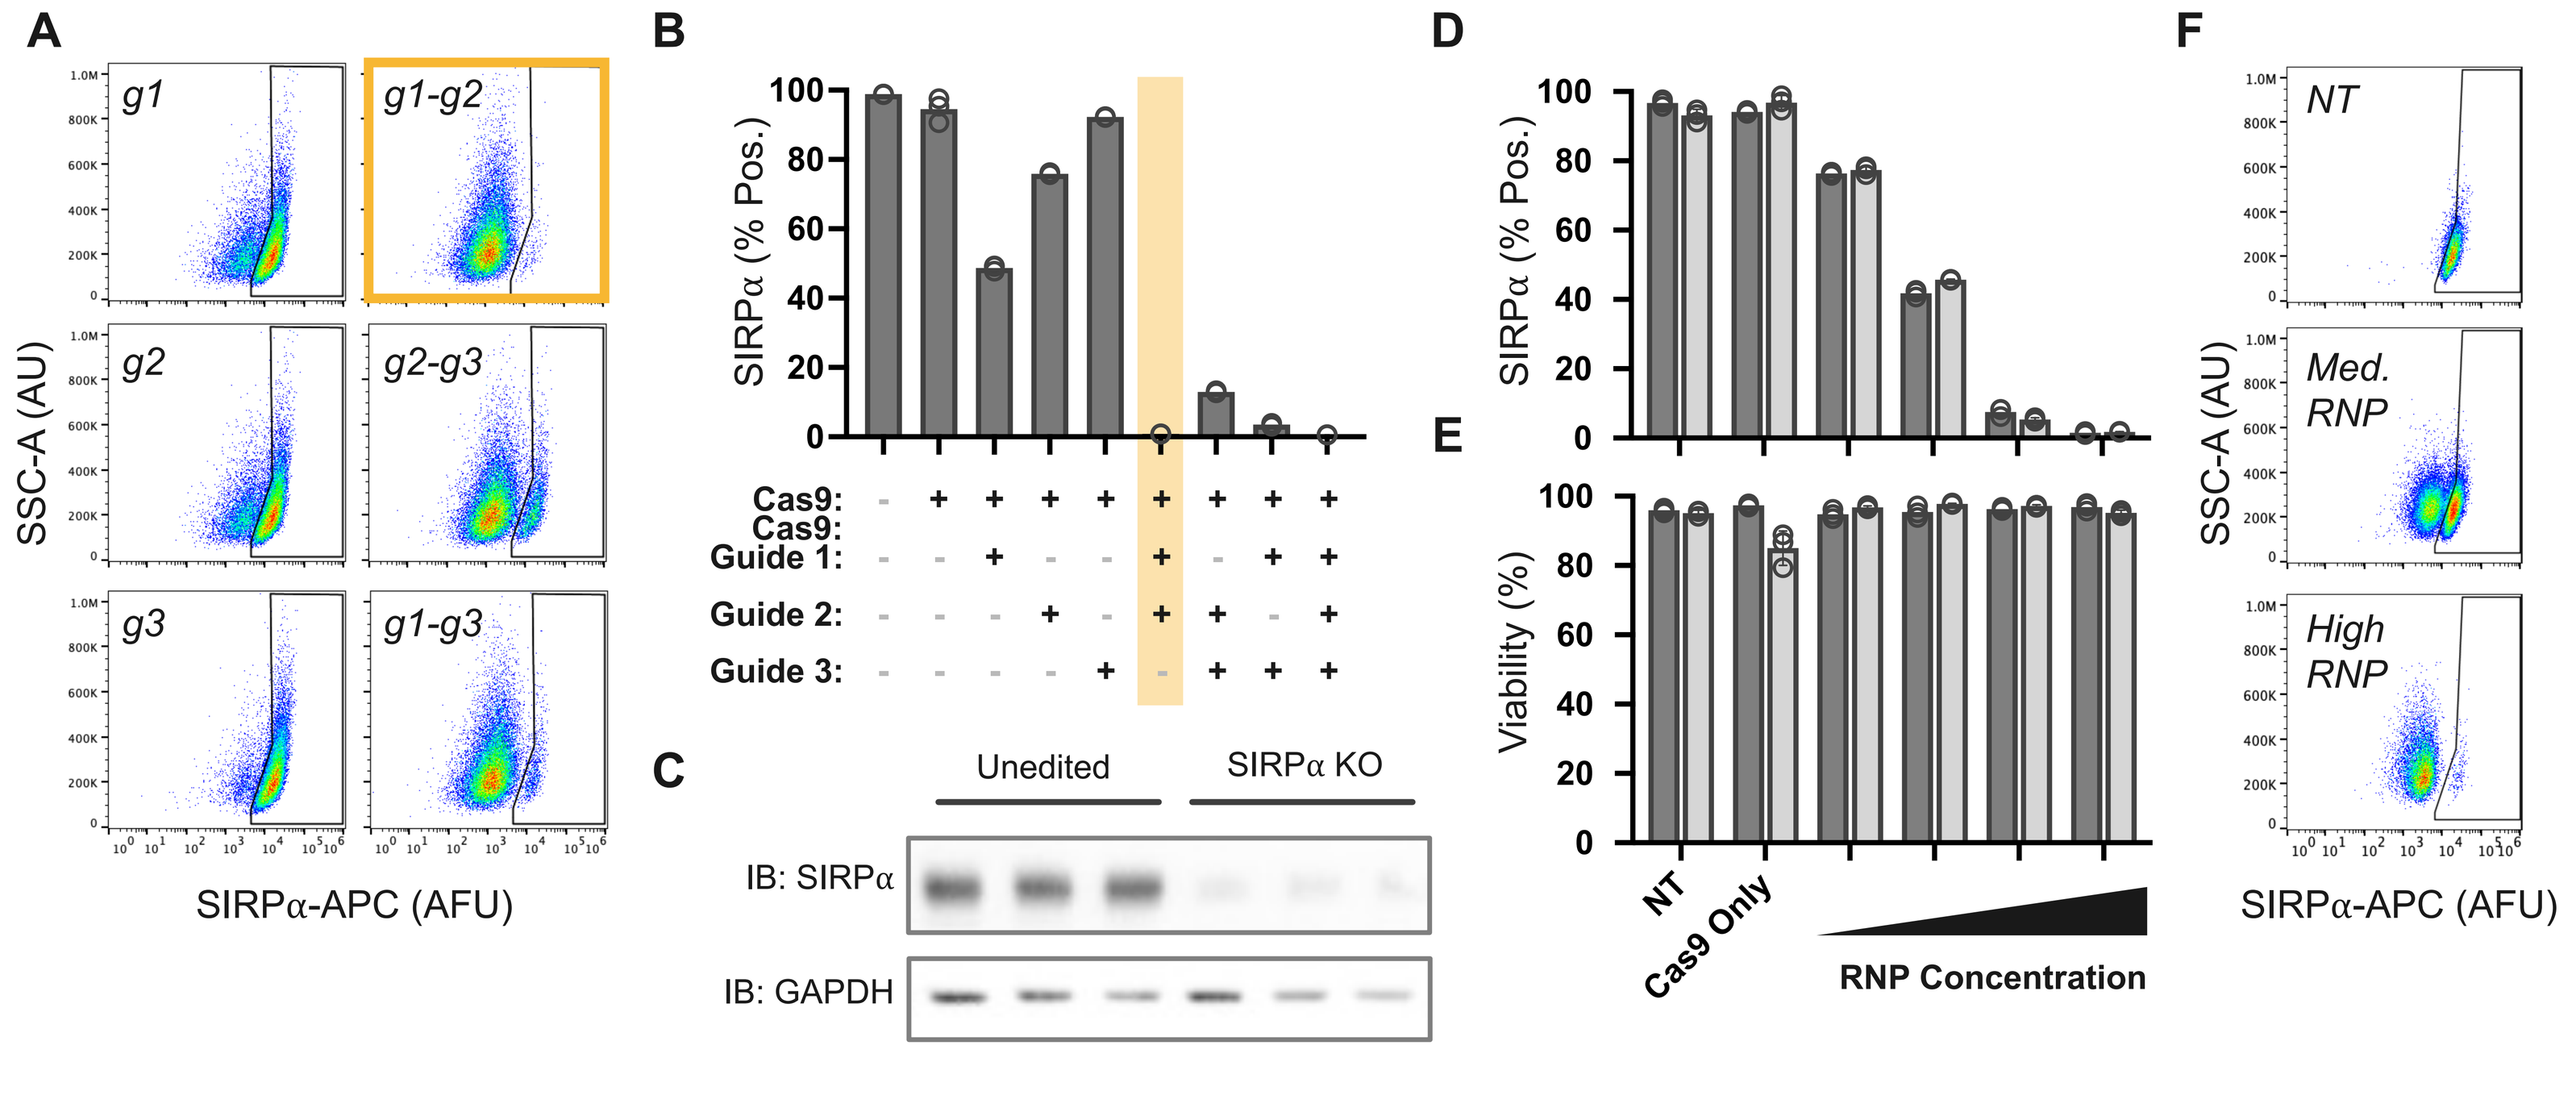
A
g1
g1-g2
g2
g2-g3
g3
g1-g3
SSC-A (AU)
SIRPα-APC (AFU)
B
SIRPα (% Pos.)
Cas9:
Guide 1:
Guide 2:
Guide 3:
C
Unedited
SIRPα KO
IB: SIRPα
IB: GAPDH
D
SIRPα (% Pos.)
E
Viability (%)
NT
Cas9 Only
RNP Concentration
F
NT
Med. RNP
High RNP
SSC-A (AU)
SIRPα-APC (AFU)

Figure 2. CRISPR/Cas9 KO of SIRPα in primary human macrophages. (**A,B**) Selection of minimal gRNA permutations needed for efficient gene KO, quantified by FCM. (**C**) Western blot evaluation of SIRPα KO. (**D-F**) SIRPα KO in macrophages from two donors (light and dark gray bars). Surface SIRPα shows gene editing is (**D**) titratable with (**E**) minimal impact on macrophage viability. (**F**) Gene KO yields distinct bimodal populations of edited and unedited macrophages. SIRPα KO in primary human macrophages is efficient and well-tolerated.

Enhanced phagocytosis and killing by SIRPα KO CAR-M

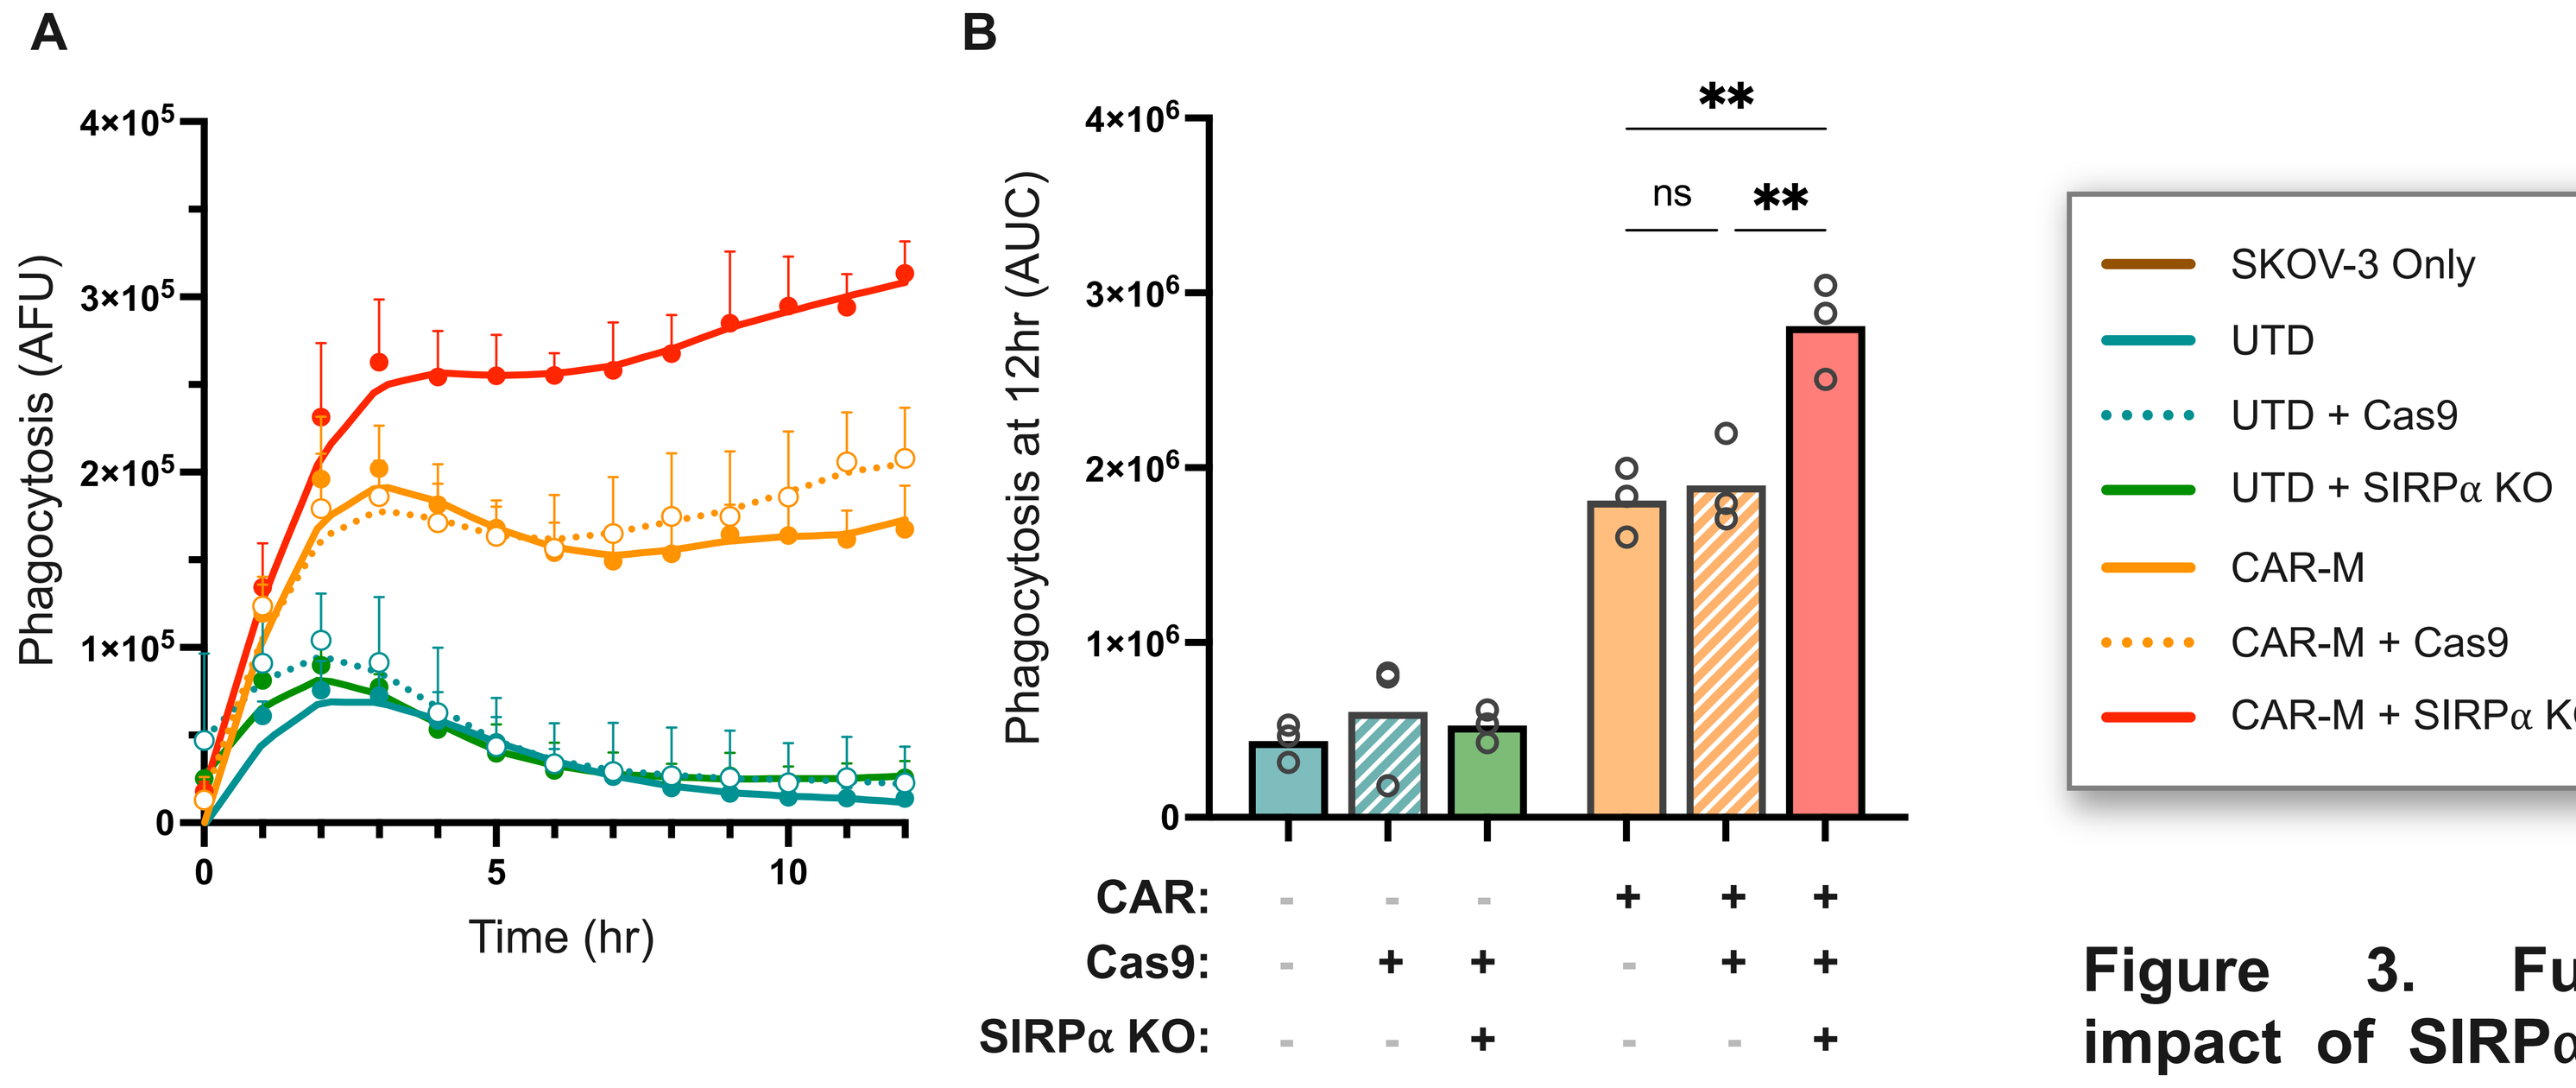
A
Phagocytosis (AFU)
Time (hr)
B
Phagocytosis at 12hr (AUC)
**
ns
**
CAR:
Cas9:
SIRPα KO:
SKOV-3 Only
UTD
UTD + Cas9
UTD + SIRPα KO
CAR-M
CAR-M + Cas9
CAR-M + SIRPα KO

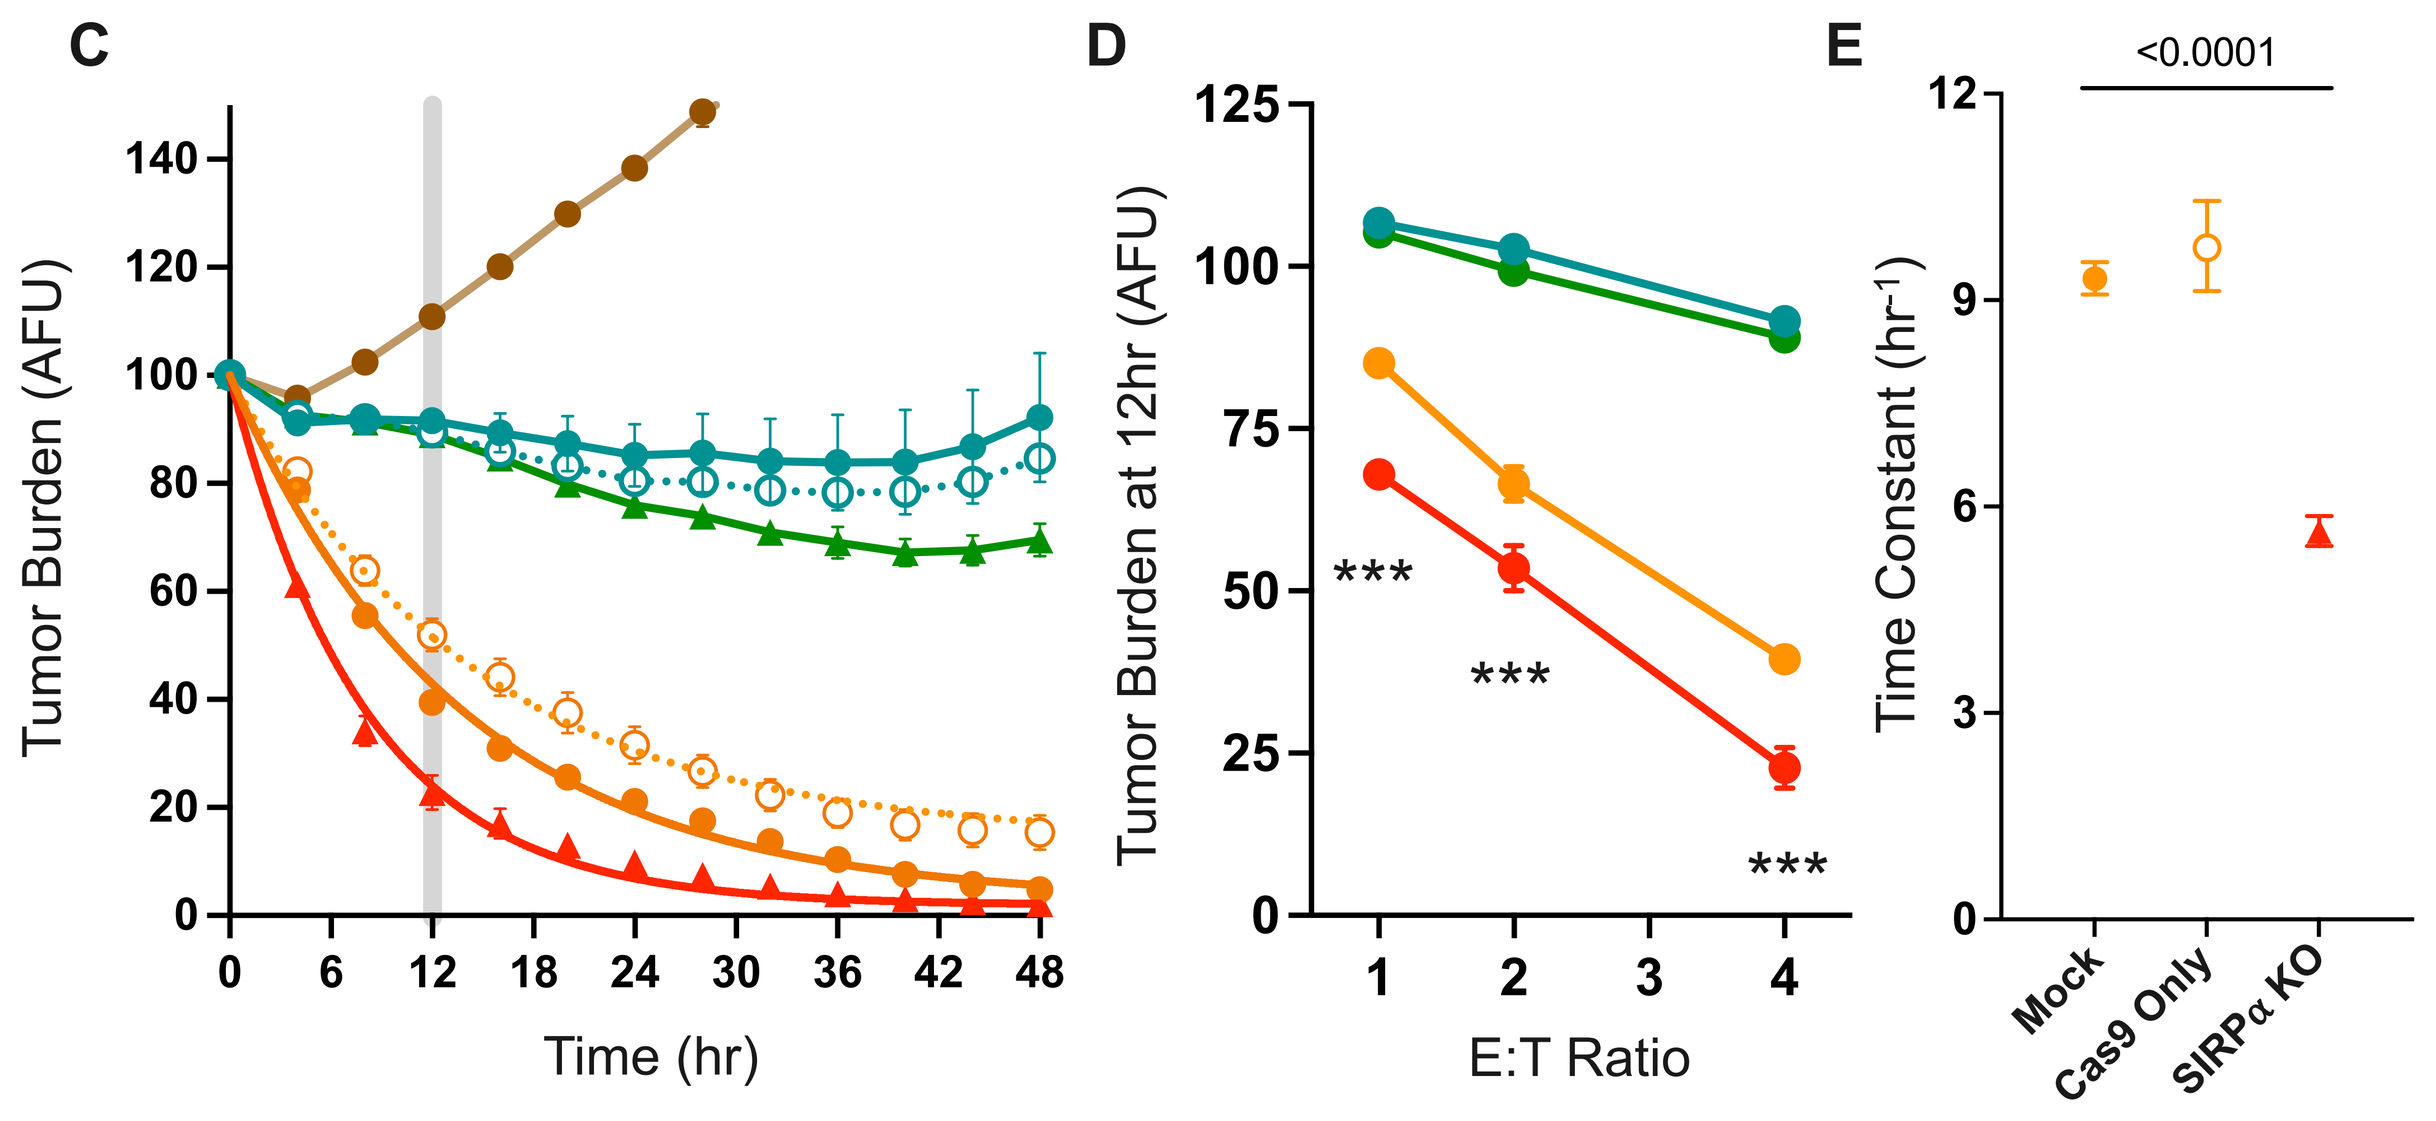
C
Tumor Burden (AFU)
Time (hr)
D
Tumor Burden at 12hr (AFU)

E:T Ratio
E
Time Constant (hr⁻¹)
<0.0001
Mock
Cas9 Only
SIRPα KO

Figure 3. Functional impact of SIRPα KO on killing and phagocytosis. (**A-B**) Phagocytic ability of macrophages cultured with SKOV-3 labeled with a pH-sensing dye that increases emission upon entering acidic phagosomes. Accumulation of signal is measured via live cell imaging. SIRPα-KO CAR-M exhibit greater phagocytic ability than unedited CAR-M. SIRPα KO in the absence of CAR transduction is not sufficient to induce phagocytic activity. (**C-E**) Growth of SKOV-3 cultured with macrophages, quantified via live cell imaging. (**C**) Unedited and SIRPα KO CAR-M kill SKOV-3 cells when co-cultured at a 4:1 E:T ratio. (**D**) Killing at 12 hr timepoint for various E:T ratios. (**E**) Time constants ± 90% CI of SKOV-3 grown with CAR-M at a 4:1 E:T ratio. SIRPα-KO CAR-M can kill target cells on shorter timescales than unedited CAR-M.
SIRPα KO in the absence of CAR transduction does not induce tumoricidal activity in macrophages.

SIRPα KO CAR-M maintain M1 polarization

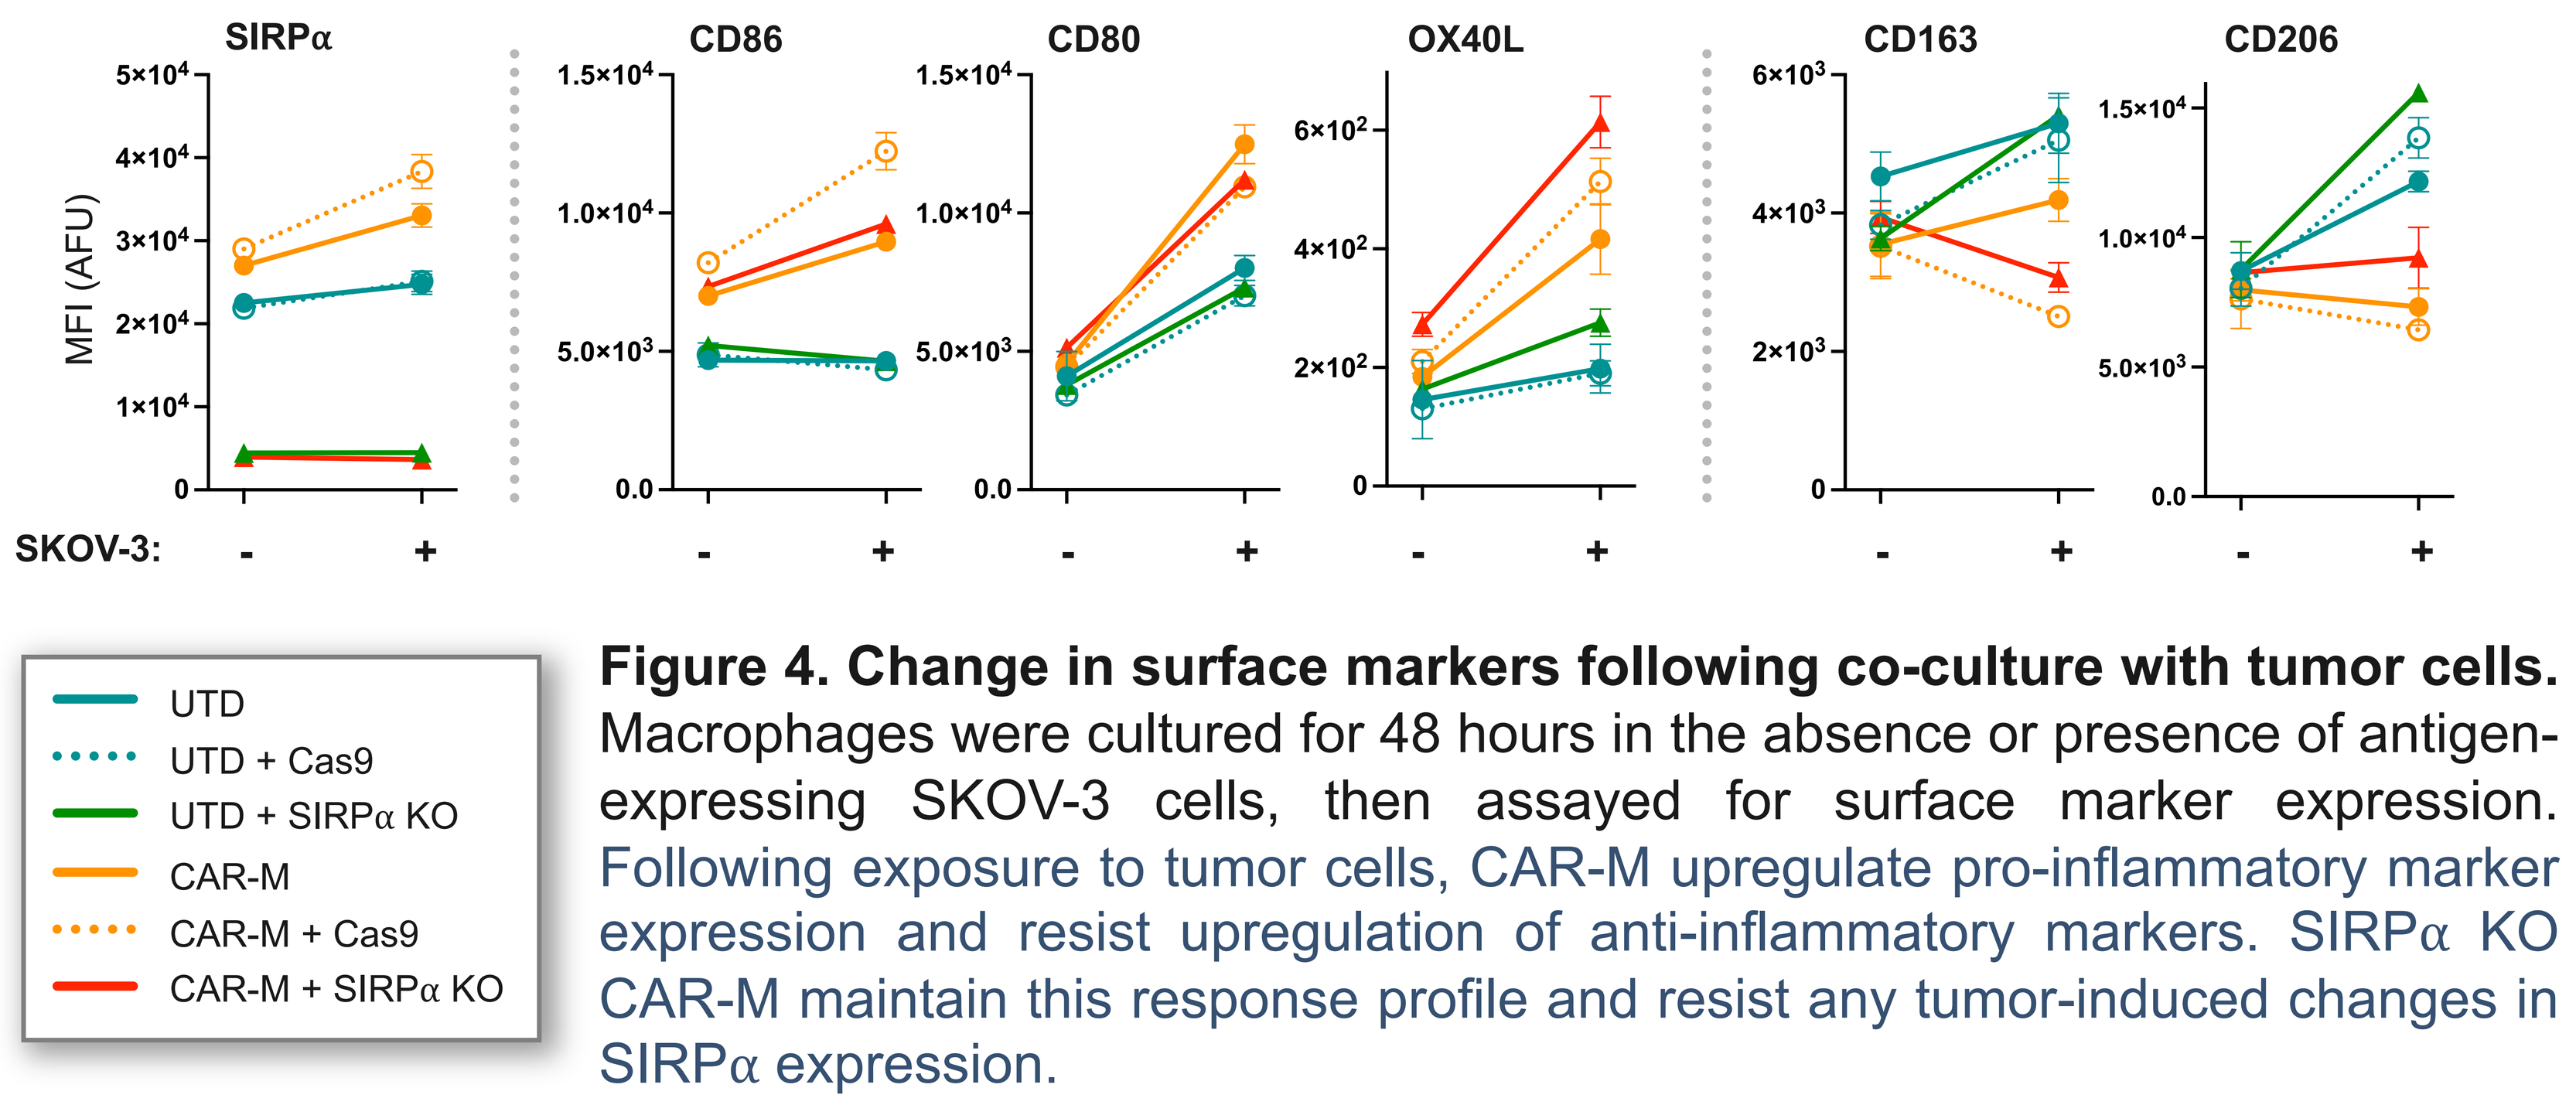
SIRPα
CD86
CD80
OX40L
CD163
CD206
MFI (AFU)
SKOV-3:
UTD
UTD + Cas9
UTD + SIRPα KO
CAR-M
CAR-M + Cas9
CAR-M + SIRPα KO

Figure 4. Change in surface markers following co-culture with tumor cells. Macrophages were cultured for 48 hours in the absence or presence of antigen-expressing SKOV-3 cells, then assayed for surface marker expression. Following exposure to tumor cells, CAR-M upregulate pro-inflammatory marker expression and resist upregulation of anti-inflammatory markers. SIRPα KO CAR-M maintain this response profile and resist any tumor-induced changes in SIRPα expression.

SIRPα-KO CAR-M BYPASS CD47 INHIBITION

SIRPα-KO CAR-M release pro-inflammatory cytokines in the presence of CD47

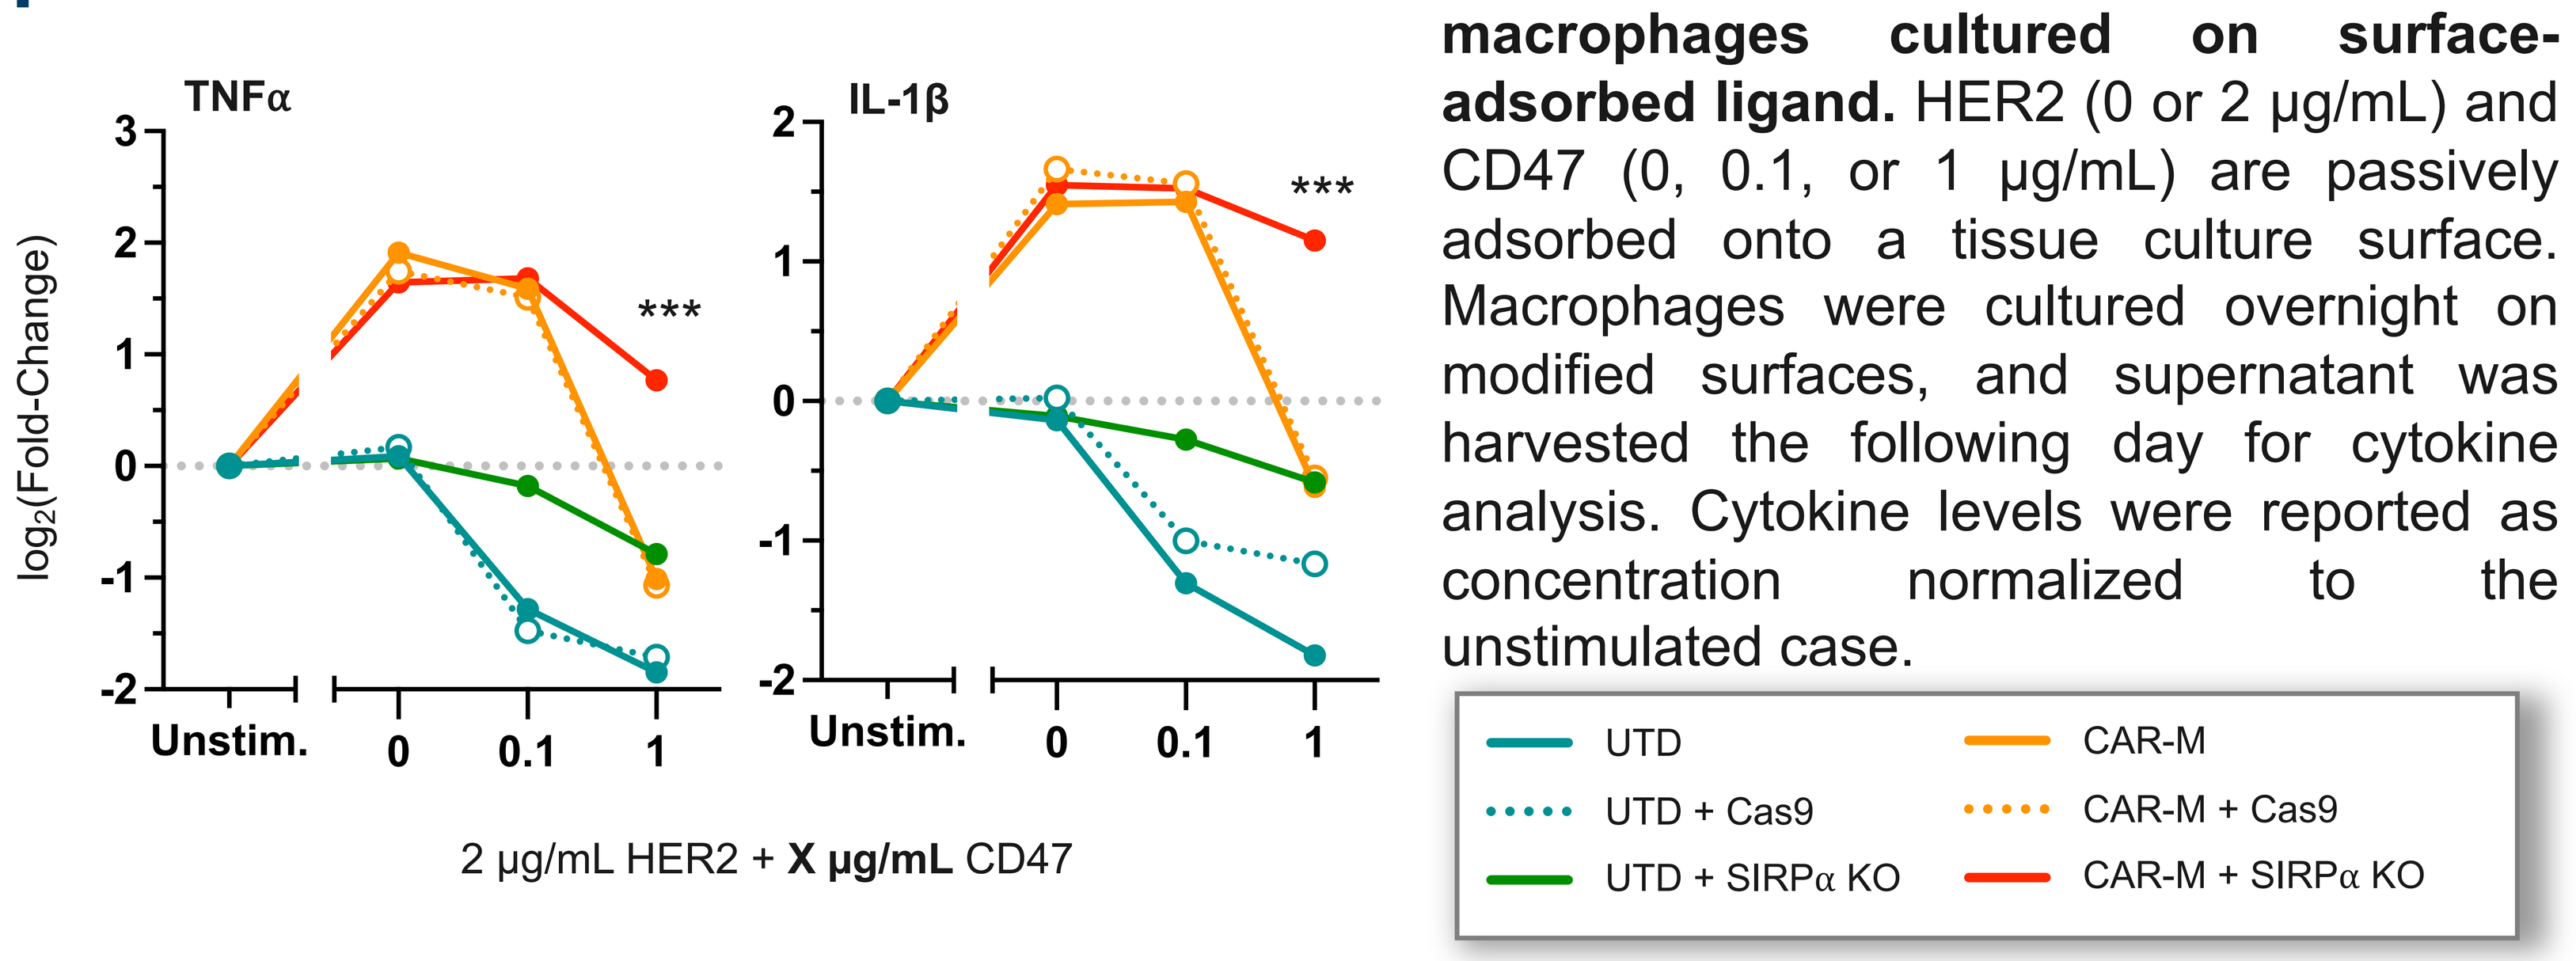
TNFα
IL-1β
log₂(Fold-Change)

Unstim.
2 µg/mL HER2 + X µg/mL CD47
UTD
UTD + Cas9
UTD + SIRPα KO
CAR-M
CAR-M + Cas9
CAR-M + SIRPα KO

Figure 5. Cytokine secretion from macrophages cultured on surface-adsorbed ligand. HER2 (0 or 2 µg/mL) and CD47 (0, 0.1, or 1 µg/mL) are passively adsorbed onto a tissue culture surface. Macrophages were cultured overnight on modified surfaces, and supernatant was harvested the following day for cytokine analysis. Cytokine levels were reported as concentration normalized to the unstimulated case.

Exposure to CD47 inhibits CAR-M cytokine secretion and reduces TNFα and IL-1β to basal expression levels. SIRPα-KO CAR-M, however, maintain elevated cytokine expression in the presence of CD47.

CONCLUSIONS

CAR-Macrophage anti-tumor activity can be enhanced by blocking checkpoint signals.

Primary human CAR-M can be efficiently edited using CRISPR/Cas9.

SIRPα-KO CAR-M exhibit increased killing and phagocytic capabilities and bypass the CD47 checkpoint.

This work provides the first example of combining primary human CAR-M with additional genetic engineering.

Ongoing studies include evaluating the ability of SIRPα-KO CAR-M to clear solid tumors and stimulate host immune cells *in vivo*.

[1] Klichinsky, M. *et al.* Human chimeric antigen receptor macrophages for cancer immunotherapy. *Nat Biotechnol. 2020.*
[2] Anderson, N. R., Minutolo, N. G., et al. Macrophage-Based Approaches for Cancer Immunotherapy. *Cancer Res.* 2021.
[3] Pierini, S. *et al.* Abstract 63: Chimeric antigen receptor macrophages (CAR-M) induce anti-tumor immunity and synergize with T cell checkpoint inhibitors in pre-clinical solid tumor models. *Immunology.* 2021.
[4] Willingham, S. B. *et al.* The CD47-signal regulatory protein alpha (SIRPa) interaction is a therapeutic target for human solid tumors. *Proc National Acad Sci.* 2012.

carisma THERAPEUTICS